Neurofilament light as a biomarker in MS: What is the current perception among UK MS specialists?

Adam Brown, PhD (Novartis)
Dean Patrick (Novartis)
Steve Medhurst, PhD (Novartis)

Background

Multiple sclerosis (MS) displays a highly variable clinical course that ranges from reversible episodes of impairment to severe disability with neuronal damage present from an early stage.[1,2] Due to the unpredictable and heterogeneous nature of both the disease course and treatment response, biomarkers reflecting these processes are in high demand.[2]

Neurofilaments are neuron-specific cytoskeletal proteins that can be released following axonal damage.[2] As such, elevated levels of neurofilament light chains (NfL) in cerebrospinal fluid (CSF) or peripheral blood are thought to reflect axonal damage and neuronal death in MS. They are emerging as a promising biomarker for the evaluation of disease activity and treatment response.[2] However, the potential benefit of NfL testing for the clinical management of MS remains uncertain.

Aim and methods

In order to assess the potential impact NfL testing could have on the clinical management of MS, 22 MS consultants and 1 MS specialist nurse based in the UK completed a 14-question survey between 1 August and 31 August 2018. The objectives of this survey were to gain insights into:

1. The current understanding of NfL as a biomarker in MS
2. How NfL relates to distinct disease processes
3. The potential role for NfL in MS disease monitoring
4. MS specialists current level of experience in measuring NfL
5. The interest in measuring NfL in patients if a test was readily available

The survey questions were structured to be answered either on a rating scale of 1 to 10 (with 1 being the lowest rating and 10 being the highest), or through a multiple choice format.

Summary and conclusions

The survey results provide an indication of how UK MS specialists view the potential impact of NfL testing in MS

The majority of participants were aware of the value of NfL as a biomarker of neuronal damage in MS, and would use a test if available

However, further clarity on how NfL levels relate to underlying disease processes is still required

In the context of clinical trials, most MS specialists thought that NfL should be evaluated as a clinical endpoint, and could be a potential target for the assessment of NEDA status

Clinical scenarios where participants felt NfL assessment could add value included for the confirmation of aggressive disease at diagnosis, to evaluate patient prognosis, to monitor disease progression and response to therapy, and for the confirmation of relapse

To build a more complete picture we now intend to survey MS nurse specialists

Barriers to the use of NfL in clinical practice

- Lack of clarity on the correlation between NfL levels and the underlying pathological processes it reflects
- Cost of serum assay and access to serum via lumbar puncture
- Clinicians lack confidence in NfL due to lack of knowledge when compared with magnetic resonance imaging (MRI)

Results

General opinion on NfL

- Among the 23 MS specialists surveyed, the mean rating for understanding NfL as a biomarker of MS was 6.6 (range 3–10)
- While the vast majority (91.3%) of participants thought that NfL levels were reflective of neuronal damage in MS, there was less consensus around:
 - the association with clinical and radiological activity (Figure 1)
 - the relation to acute inflammatory processes (Figure 2)
 - the predictive value for disease progression (Figure 2)
- Nine of the 23 MS specialists surveyed (39.1%) indicated that they already had experience with or currently measured NfL levels in their clinical practice

NfL in clinical trials

- The vast majority (91.3%) of participants thought that NfL levels should be monitored in clinical trials, with the potential of NfL as a target to establish no evidence of disease activity (NEDA) status highlighted (Figure 3)

NfL in clinical practice

- The participants thought that NfL level measurement would add value to clinical and MRI monitoring in MS, and could be a useful measure of subclinical activity (Figure 4)
- If a test was available, 95.6% of participants would be interested in measuring CSF or blood NfL levels in their patients
- Specific patient groups and clinical situations where the participants would measure NfL levels are highlighted in Figures 5 and 6, respectively

Figure 1 | **Do NfL levels reflect clinical and/or radiological activity?**

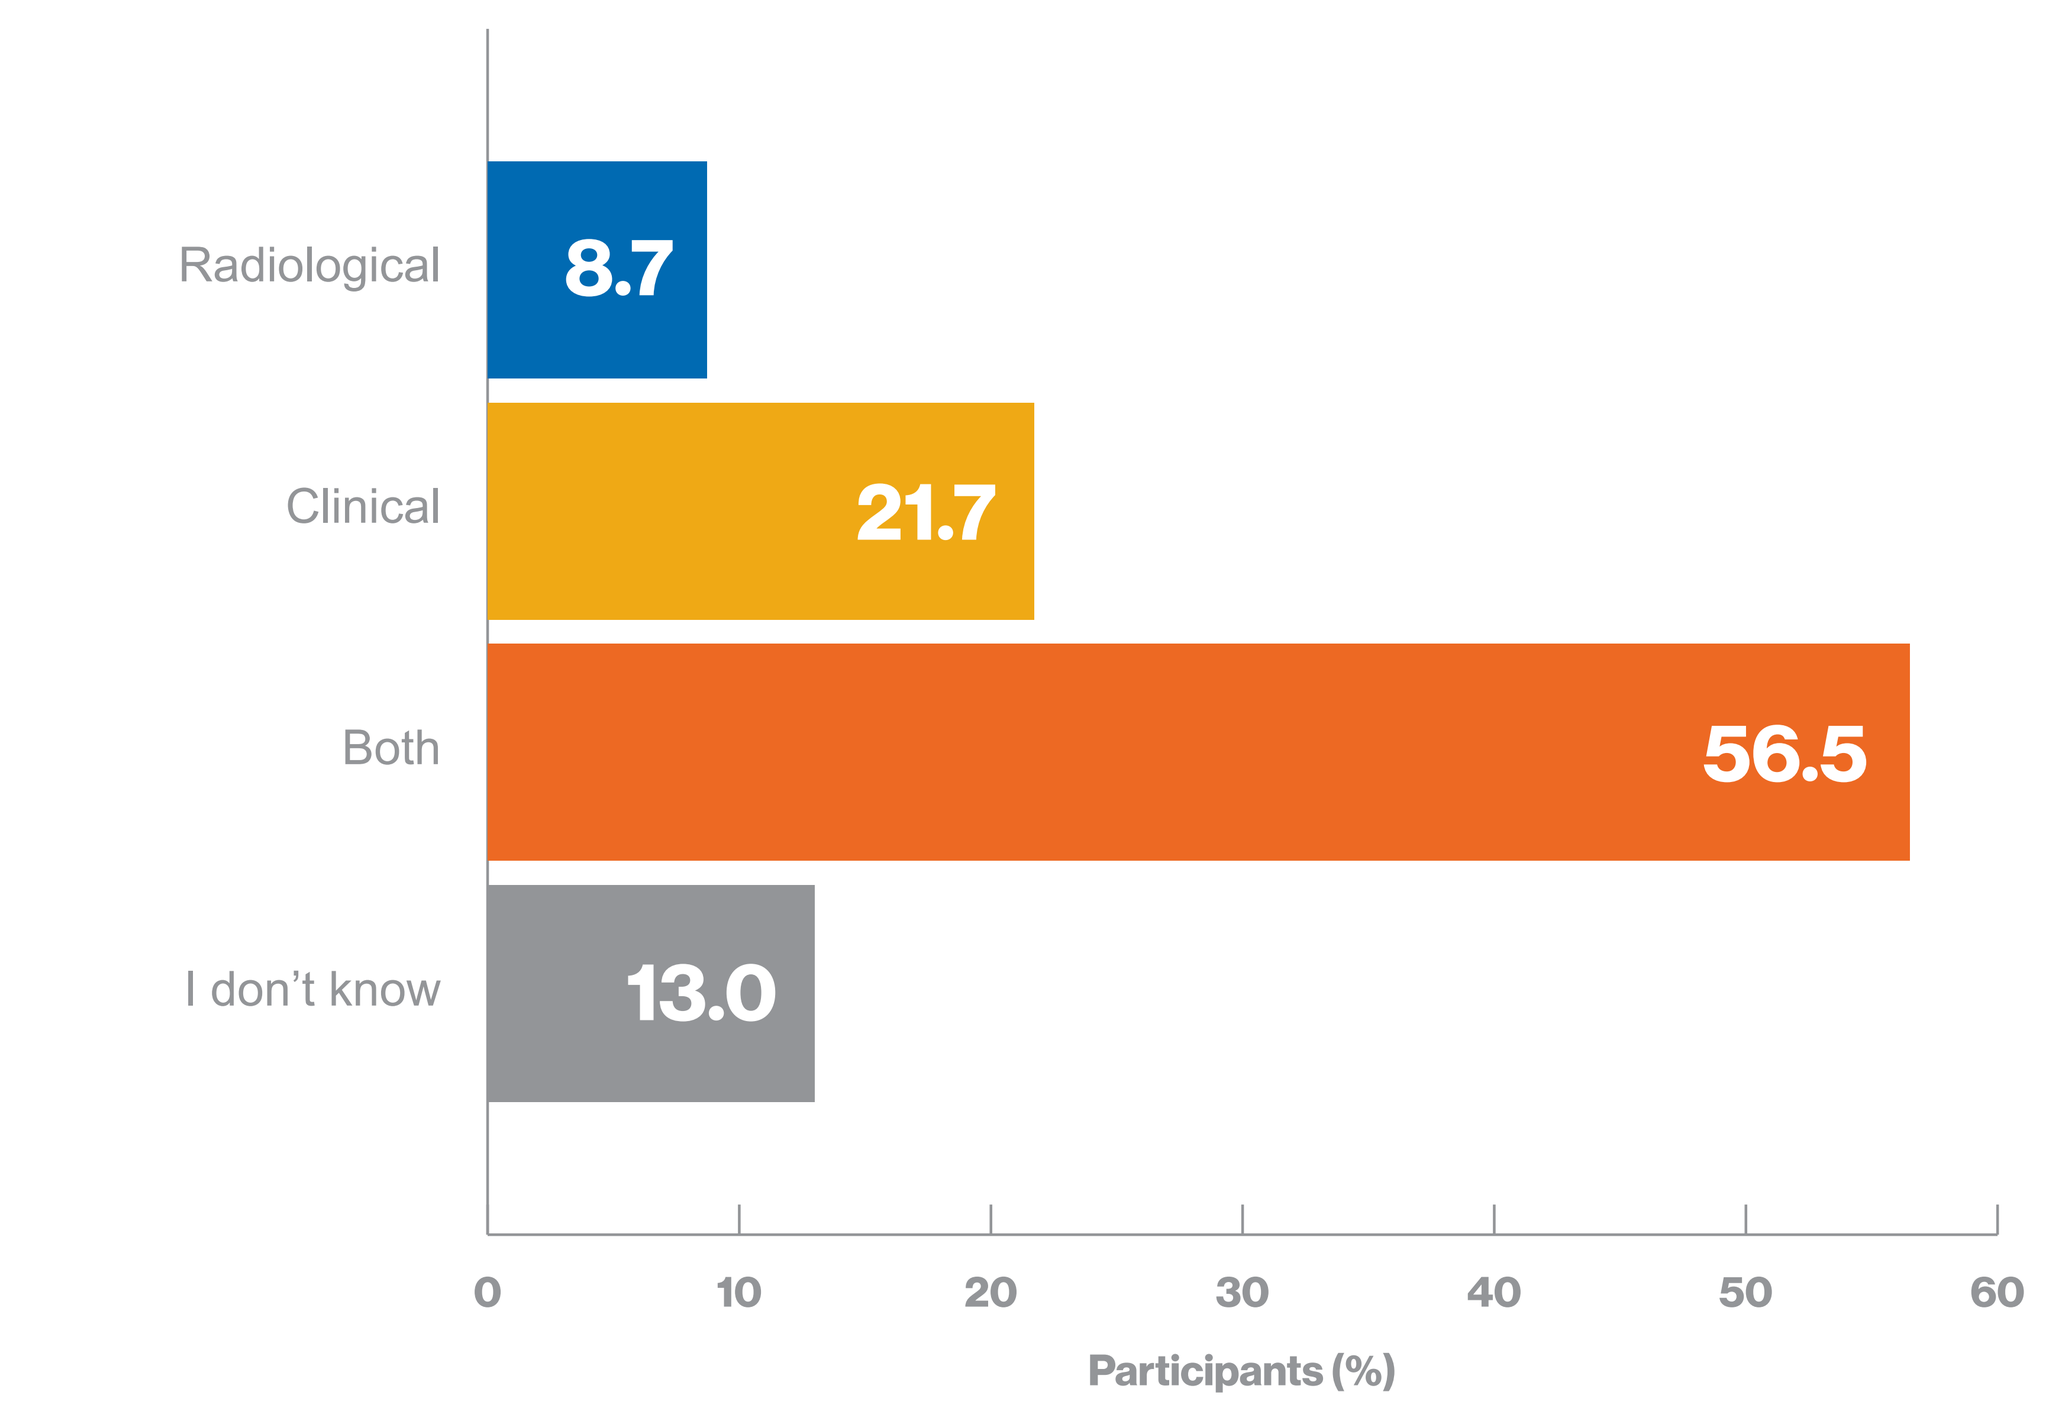
Radiological
8.7
Clinical
21.7
Both
56.5
I don't know
13.0
0
10
20
30
40
50
60
Participants (%)

Figure 2 | **Do NfL levels only reflect acute inflammatory processes or are they predictive of disability progression?**

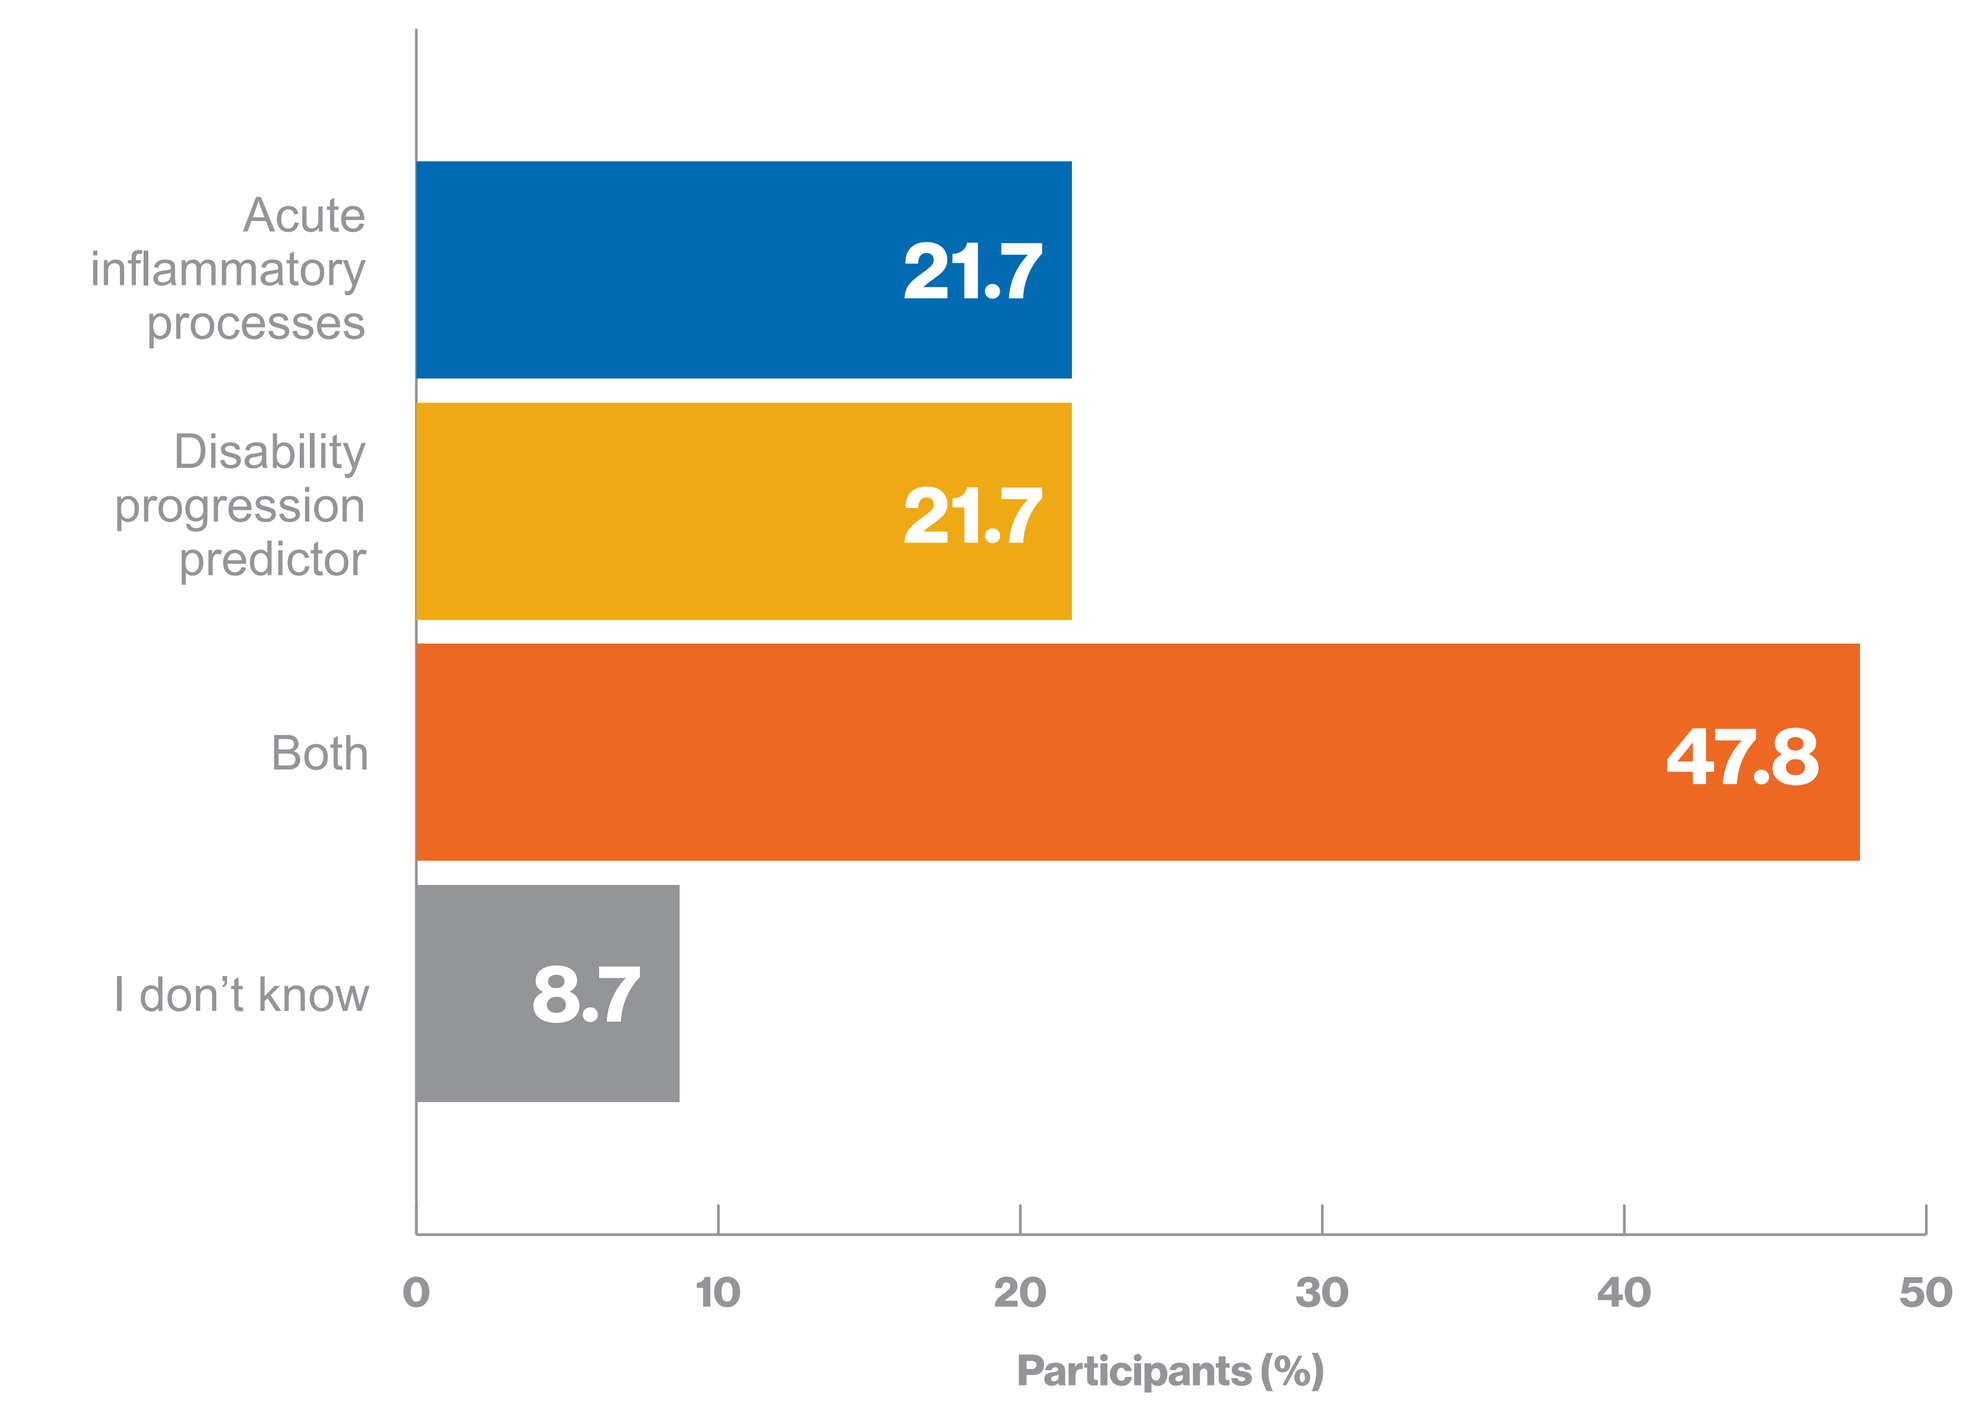
Acute inflammatory processes
21.7
Disability progression predictor
21.7
Both
47.8
I don't know
8.7
0
10
20
30
40
50
Participants (%)

Figure 3 | **Could NfL levels be integrated in a target of NEDA in clinical trials as a marker of neurodegeneration?**

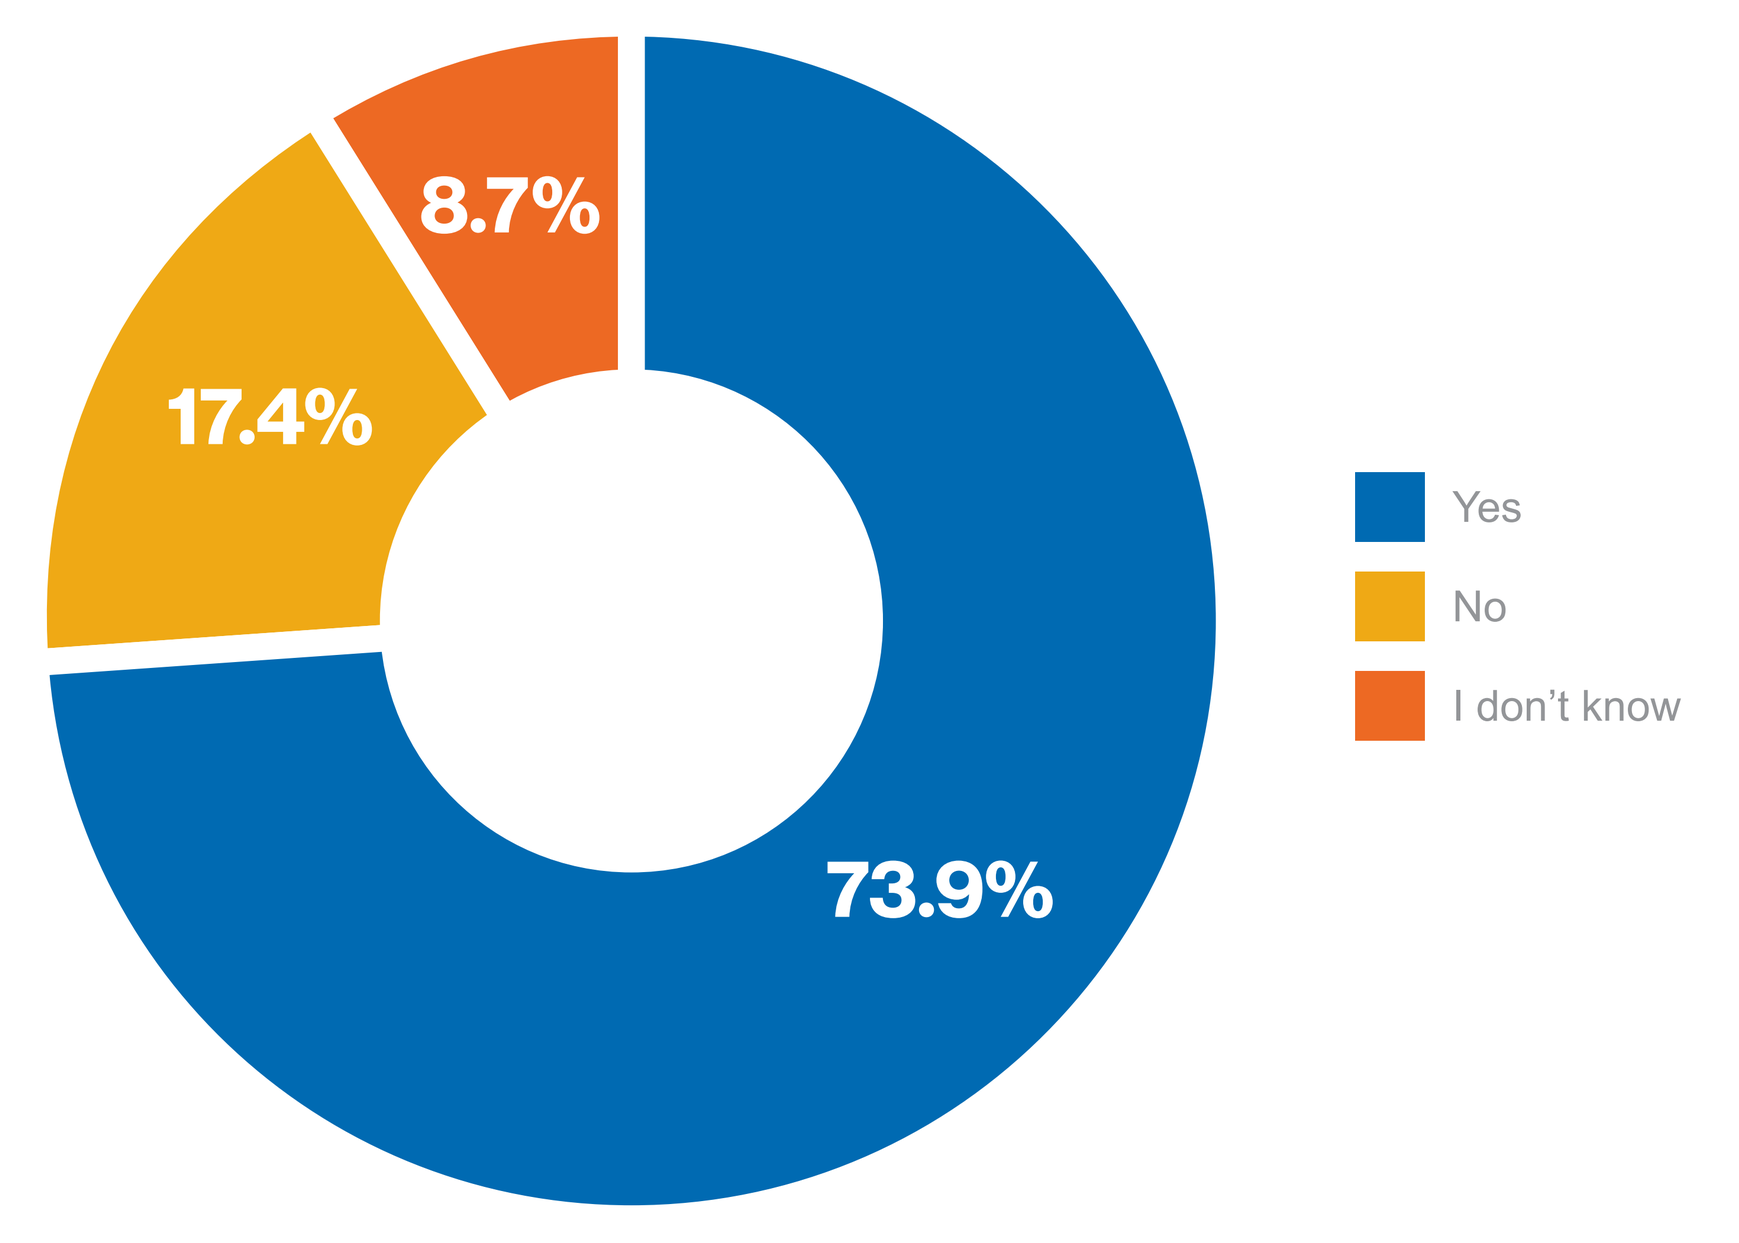
73.9%
17.4%
8.7%
Yes
No
I don't know

Figure 4 | **Survey participants rating of the value and utility of NfL level measurement**

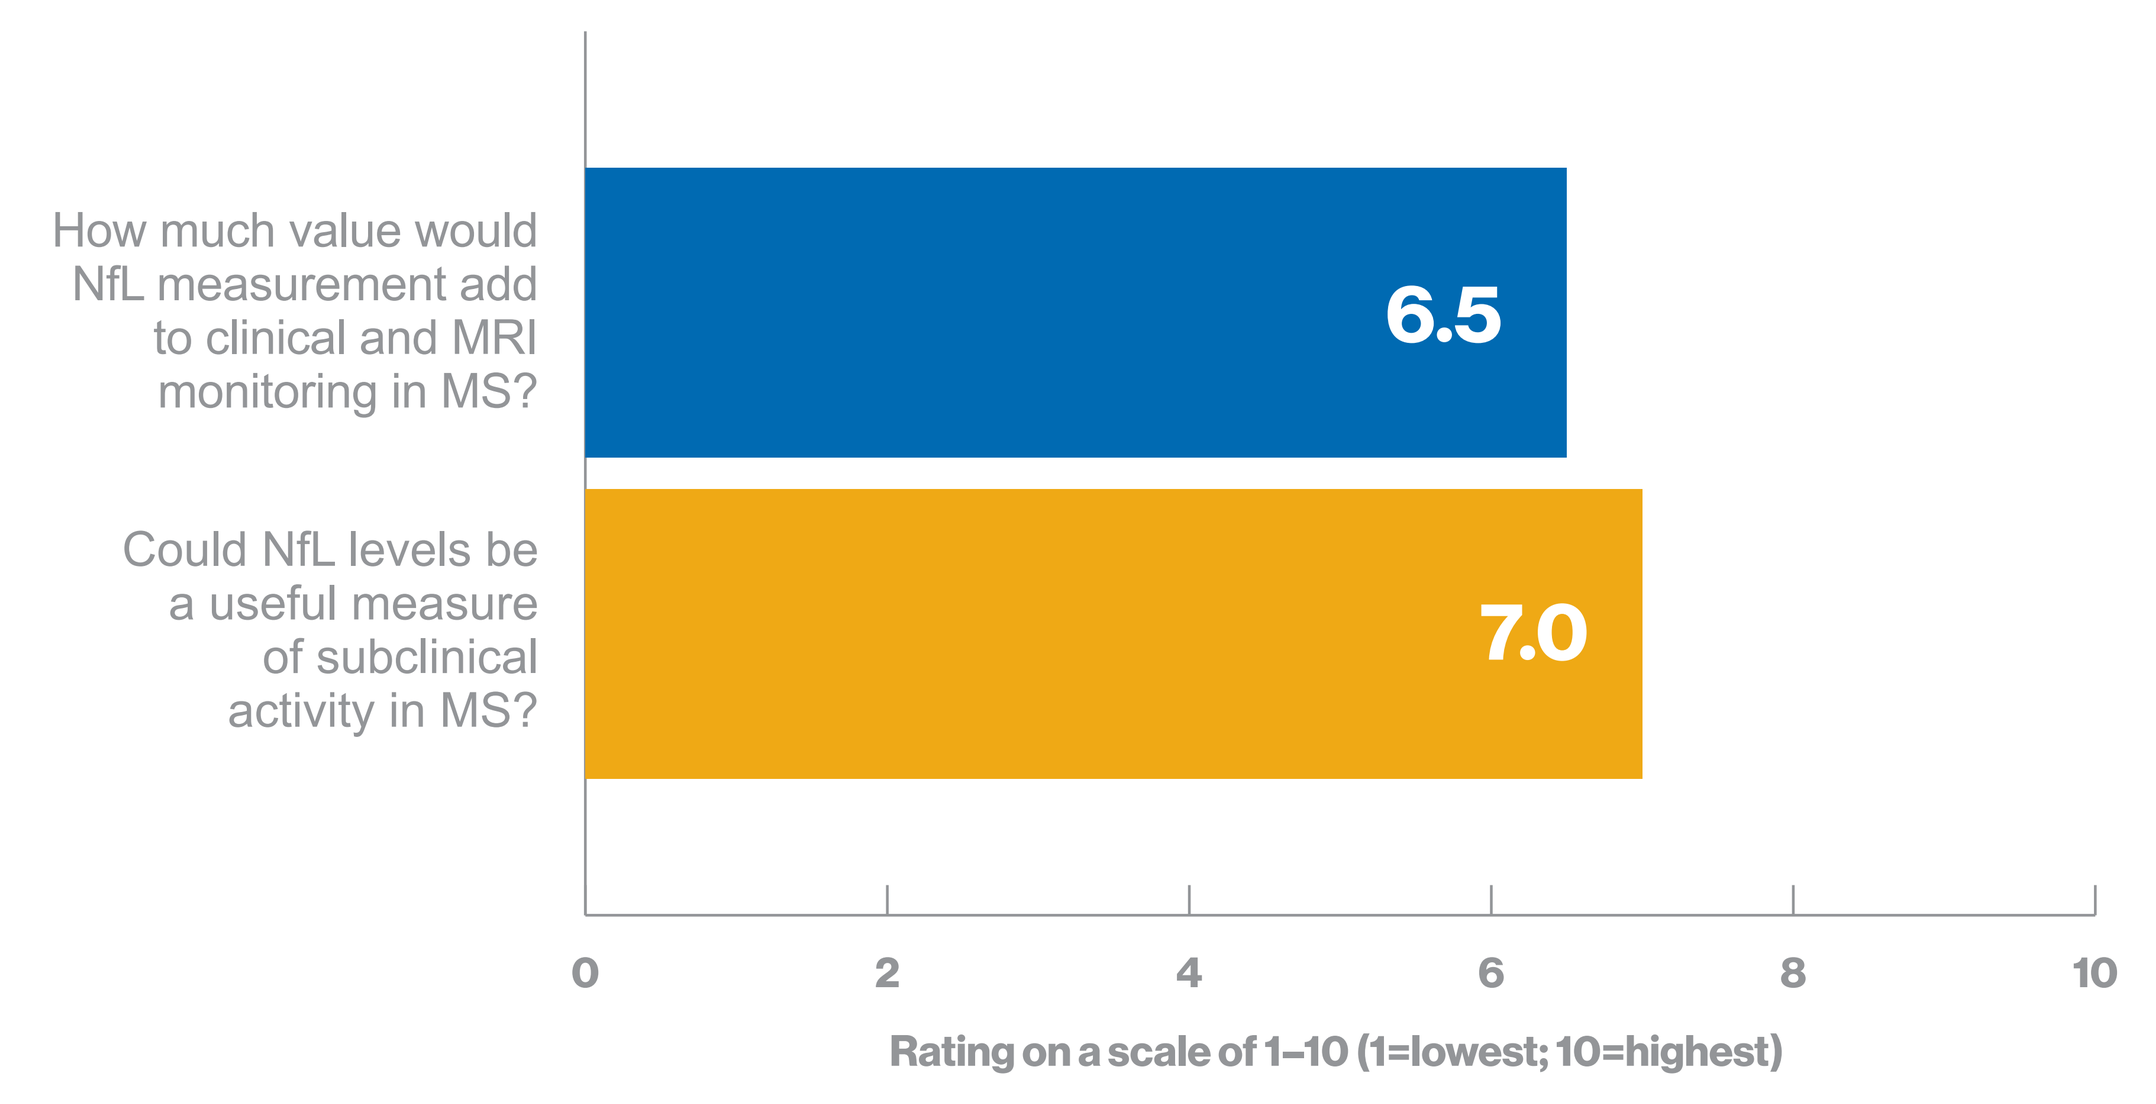
How much value would NfL measurement add to clinical and MRI monitoring in MS?
6.5
Could NfL levels be a useful measure of subclinical activity in MS?
7.0
0
2
4
6
8
10
Rating on a scale of 1–10 (1=lowest; 10=highest)

Figure 5 | **If a test was available today, in which patient groups would you measure NfL levels?**

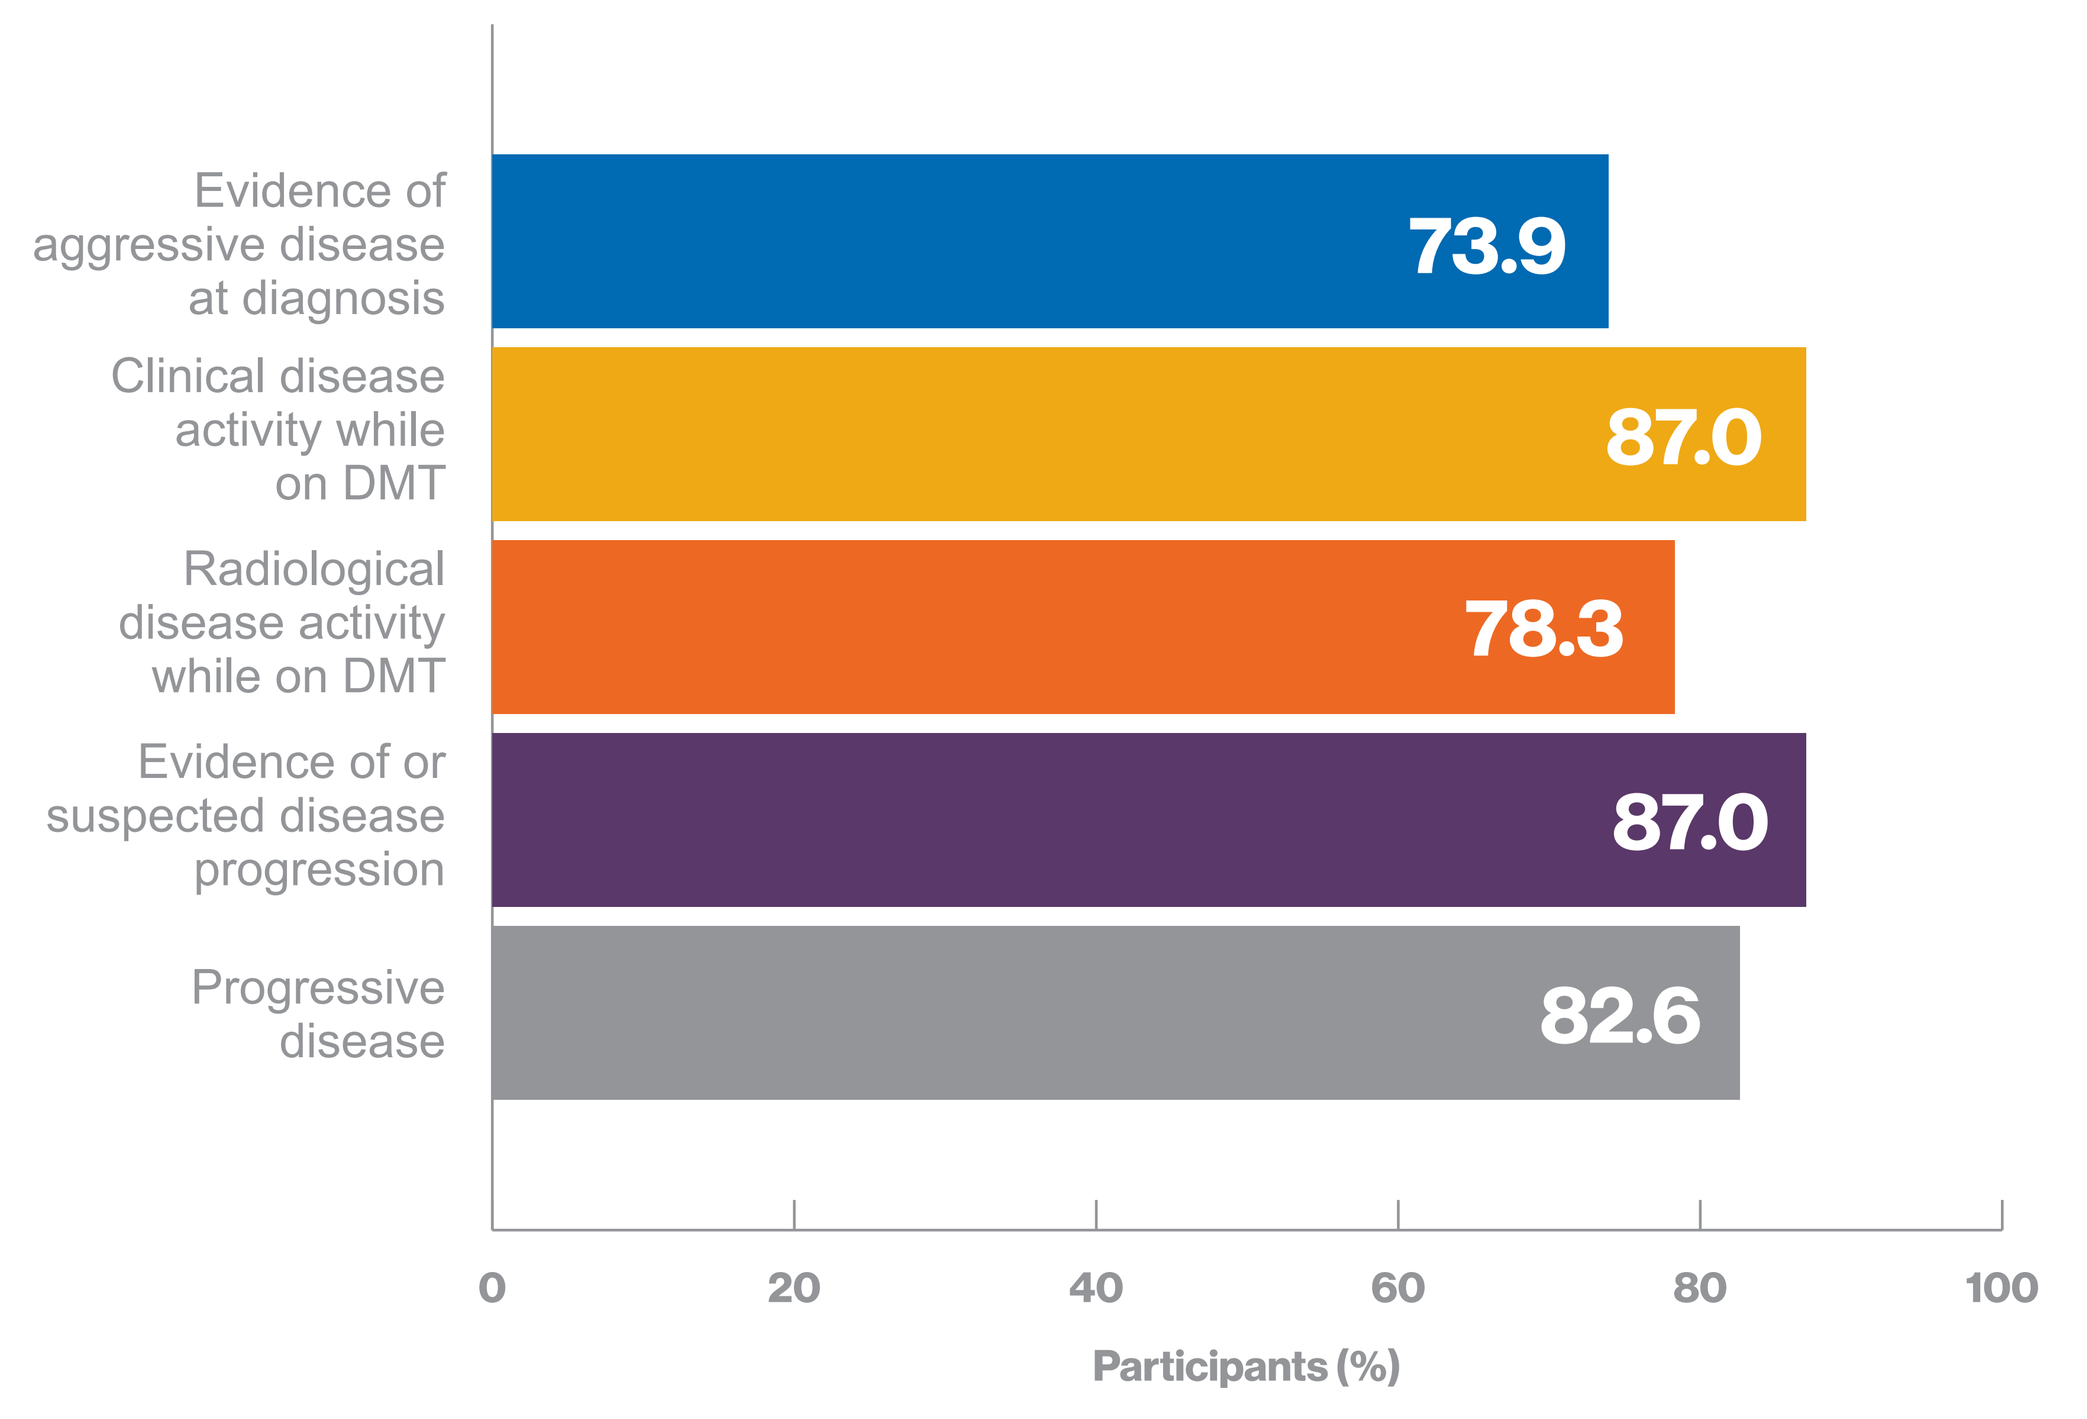
Evidence of aggressive disease at diagnosis
73.9
Clinical disease activity while on DMT
87.0
Radiological disease activity while on DMT
78.3
Evidence of or suspected disease progression
87.0
Progressive disease
82.6
0
20
40
60
80
100
Participants (%)

Figure 6 | **If a test was available today, in which of the following situations would you measure NfL levels?**

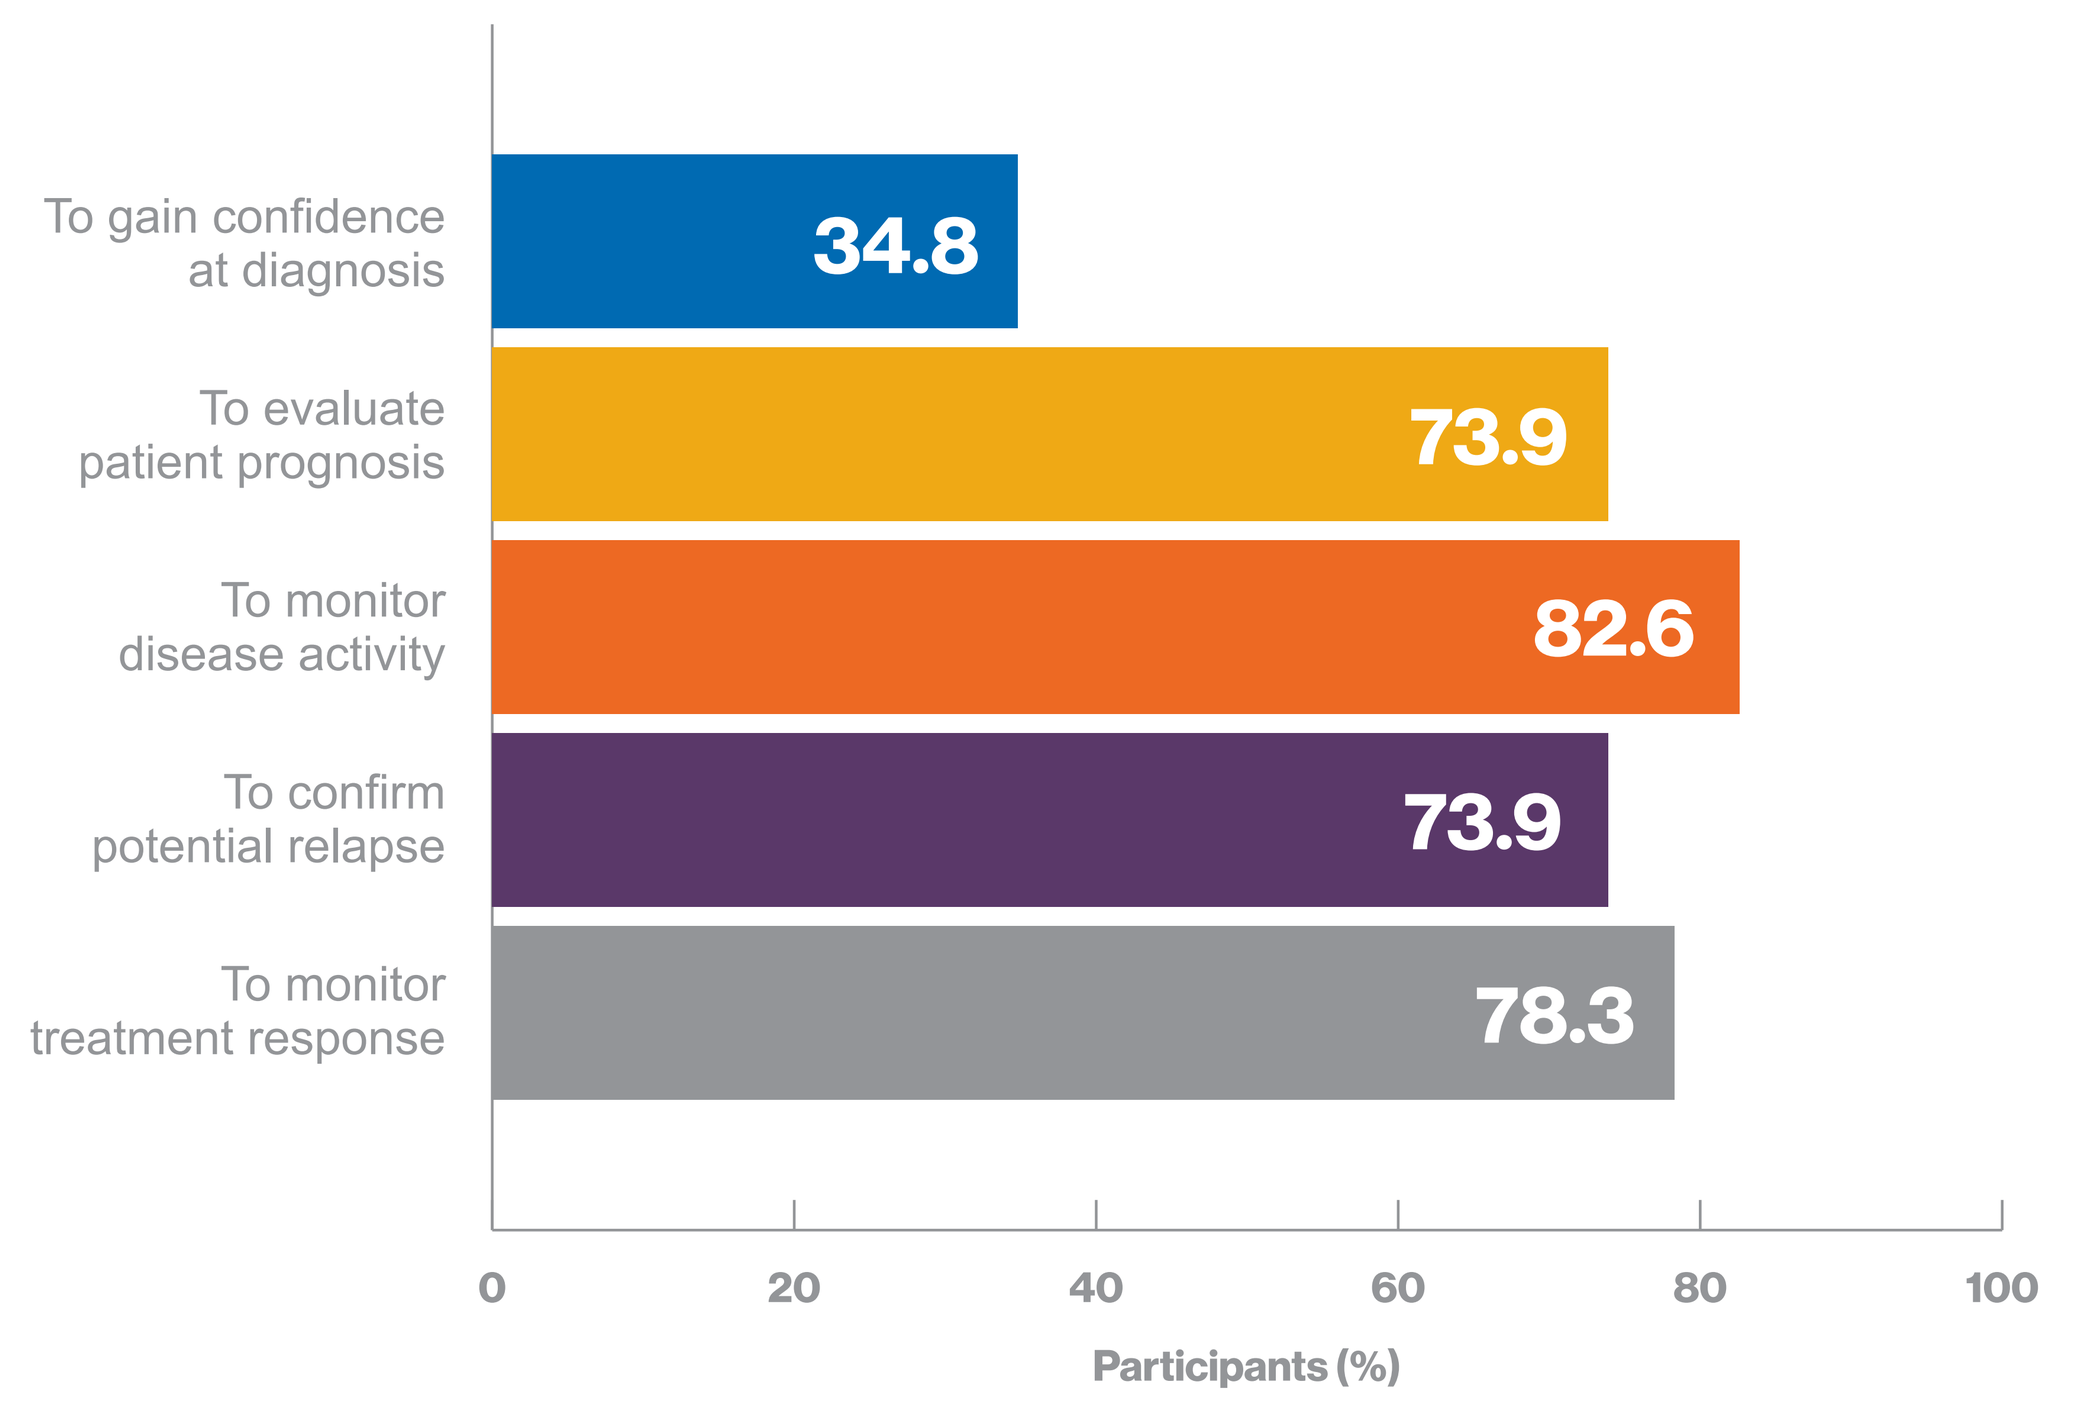
To gain confidence at diagnosis
34.8
To evaluate patient prognosis
73.9
To monitor disease activity
82.6
To confirm potential relapse
73.9
To monitor treatment response
78.3
0
20
40
60
80
100
Participants (%)

References **1.** Disanto G, et al. *Ann Neurol* 2017;81:857–870; **2.** Varhaug KN, et al. *Neurol Neuroimmunol Neuroinflamm* 2017;5:e422.
Acknowledgements On behalf of the authors we would like to acknowledge all survey participants.
Funding This survey was wholly funded by Novartis Pharmaceuticals UK Ltd. Editorial support was provided by Virgo Health, which was funded by Novartis Pharmaceuticals UK Ltd.
Abbreviations CSF=cerebrospinal fluid; DMT=disease-modifying therapy; MRI=magnetic resonance imaging; MS=multiple sclerosis; NEDA=No Evidence of Disease Activity; NfL=neurofilament light.